A Phase 1 Randomized, Double-Blind, Placebo-Controlled, Multiple Ascending Dose Study to Evaluate Safety, Tolerability and Pharmacokinetics of the Liver-Selective TR-Beta Agonist VK2809 (MB07811) in Hypercholesterolemic Subjects. B. Lian, R. Hanley and S. Schoenfeld, Viking Therapeutics, San Diego, CA

Abstract

Background: Thyroid hormone is a key regulator of lipoprotein metabolism, particularly acting via the beta isoform of nuclear T3 receptor in liver. VK2809 is a small molecule prodrug of a potent thyroid beta agonist. VK2809 is selectively cleaved in hepatic tissue by the action of cytochrome P450 isozyme 3A4, to release its pharmacologically active metabolite. In animals, VK2809 has demonstrated robust cholesterol- and triglyceride-lowering properties, as well as significant reductions in liver fat content. In humans, a prior single-ascending dose study demonstrated encouraging safety and tolerability. We here report the results of 14 days of treatment in humans with mild hypercholesterolemia.

Methods: A Phase 1 study was conducted to evaluate the safety, tolerability and pharmacokinetics of VK2809 in 56 subjects with elevated serum cholesterol. Subjects were randomized to receive once-daily oral VK2809 doses of 0.25, 1.0, 2.5, 5.0, 10, 20, and 40 mg, or placebo for 14 days.

Results: Exposures of VK2809 and the active metabolite each increased in a dose-proportional manner up to 40 mg. The terminal half-life of the active metabolite ranged from 13–41 hours and was longer at higher doses. Less than 3% of VK2809 and its active metabolite were excreted renally.

VK2809 was found to be safe and well-tolerated in this study. No treatment- or dose-related trends were observed for abnormal vital signs, ECGs, Holter monitored cardiac rhythm, or physical examination assessments. No SAEs were reported. Mean ALT levels increased with increasing doses and reached a maximum of 1.5x ULN in the 40 mg cohort. Mild dose-dependent reductions in mean TSH were observed, but without symptoms or signs of thyroid dysfunction.

Substantial and clinically meaningful placebo-adjusted reductions in LDL-C, up to 41.2% ($p<0.05$); triglycerides, up to 78.6% ($p<0.05$); and non-HDL-C, up to 44.2% ($p<0.05$), were observed at doses of 5.0 mg and above. In addition, significant reductions in Lp(a) and Apo-B were observed. No meaningful trends in HDL-C were observed.

Conclusion: Selective activation of thyroid beta with the small molecule VK2809 may represent an attractive approach to the treatment of hypercholesterolemia.

Introduction

Thyroid hormone is a key regulator of lipoprotein metabolism, particularly acting via the beta isoform of nuclear T3 receptor in liver. The effects of thyroid hormone action on lipid metabolism include increased expression of the LDL receptor, increased cholesterol disposal through bile acid synthesis via increased expression of the enzyme CYP7A1, and reducted expression of hepatic sterol regulatory element binding protein-1 (SREBP-1). These effects suggest potential therapeutic benefits in patients with elevated LDL cholesterol and triglycerides.

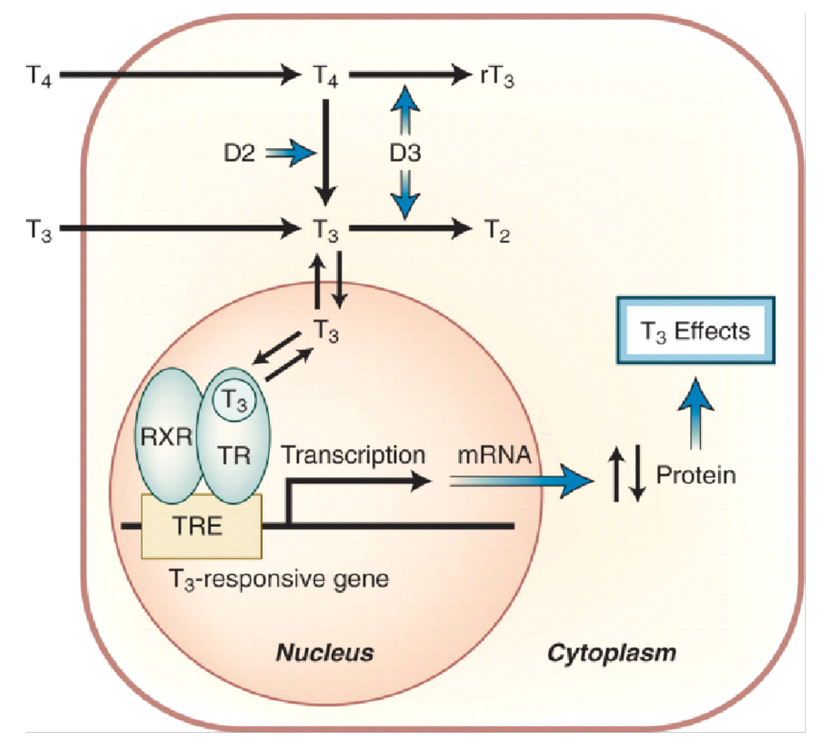

T4
T4
rT3
D2
D3
T3
T3
T2
T3
RXR
TR
Transcription
mRNA
Protein
T3 Effects
TRE
T3-responsive gene
Nucleus
Cytoplasm

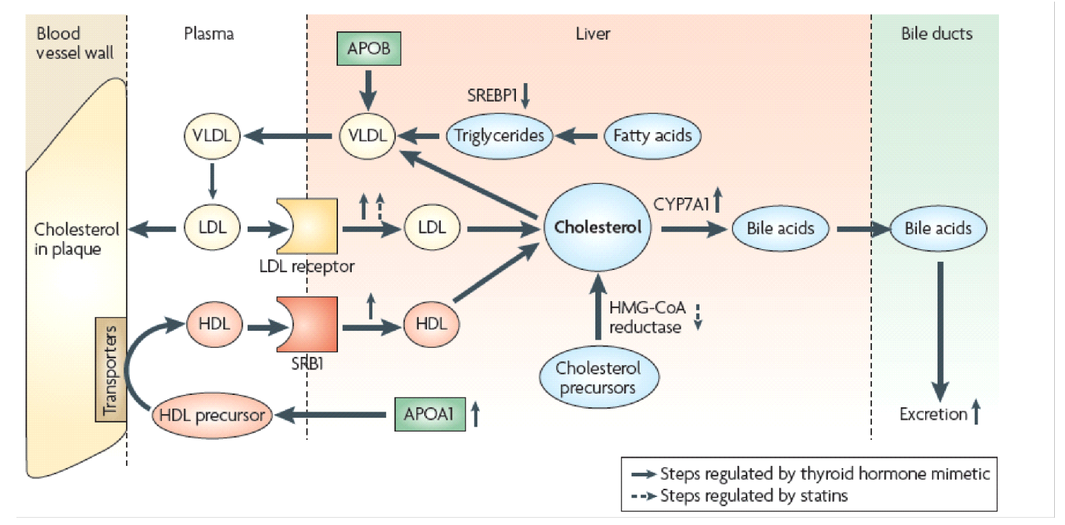

Blood vessel wall
Plasma
Liver
Bile ducts
APOB
SREBP1
VLDL
Triglycerides
Fatty acids
Cholesterol in plaque
LDL
LDL receptor
Cholesterol
CYP7A1
Bile acids
HDL
SRBI
HMG-CoA reductase
Cholesterol precursors
HDL precursor
APOA1
Excretion
Steps regulated by thyroid hormone mimetic
Steps regulated by statins

VK2809 is a small molecule prodrug of a potent thyroid beta agonist. VK2809 is selectively cleaved in hepatic tissue by the action of cytochrome P450 isozyme 3A4, to release its pharmacologically active metabolite. In animals, VK2809 has demonstrated robust cholesterol- and triglyceride-lowering properties, as well as significant reductions in liver fat content. In humans, a prior single-ascending dose study demonstrated encouraging safety and tolerability. We here report the results of 14 days of treatment in humans with mild hypercholesterolemia.

Methods

A single-center, randomized, double-blind, placebo-controlled, rising multiple-dose Phase 1 study was conducted to evaluate the safety, tolerability and pharmacokinetics of VK2809 in 56 subjects with elevated serum cholesterol. Subjects 18-54 years of age were randomized to receive once-daily oral VK2809 doses of 0.25, 1.0, 2.5, 5.0, 10, 20, or 40 mg, or placebo for 14 days. Subjects were followed for an additional 7 days off drug for safety assessment.

Overview of General Study Design

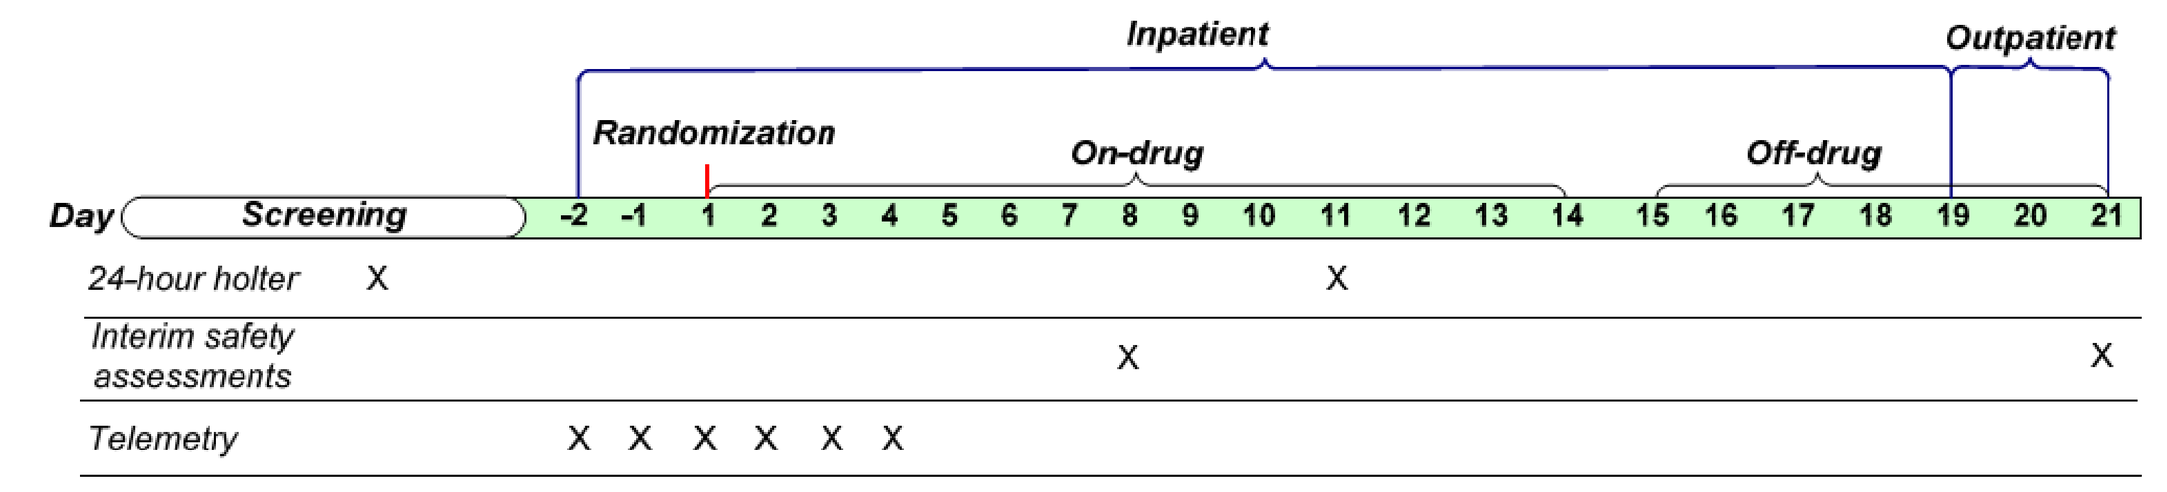

Inpatient
Outpatient
Randomization
On-drug
Off-drug
Day
Screening
-2 -1 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21
24-hour holter X X
Interim safety assessments X X
Telemetry X X X X X X

Objectives

- To characterize the pharmacokinetics (PK) of VK2809 and its active metabolite, VK2809A
- To make a preliminary assessment of LDL-C lowering by VK2809
- To assess the safety and tolerability of VK2809 in subjects with elevated cholesterol

Results

Pharmacokinetics

Plasma concentrations of VK2809 and its active metabolite VK2809A each increased in a dose-proportional manner up to 40 mg. The terminal half-life of the active metabolite ranged from 13–41 hours and was longer at higher doses. Less than 3% of VK2809 and VK2809A was excreted renally.

VK2809 and VK2809A Plasma concentration versus Time Following Multiple Doses (Day 14)

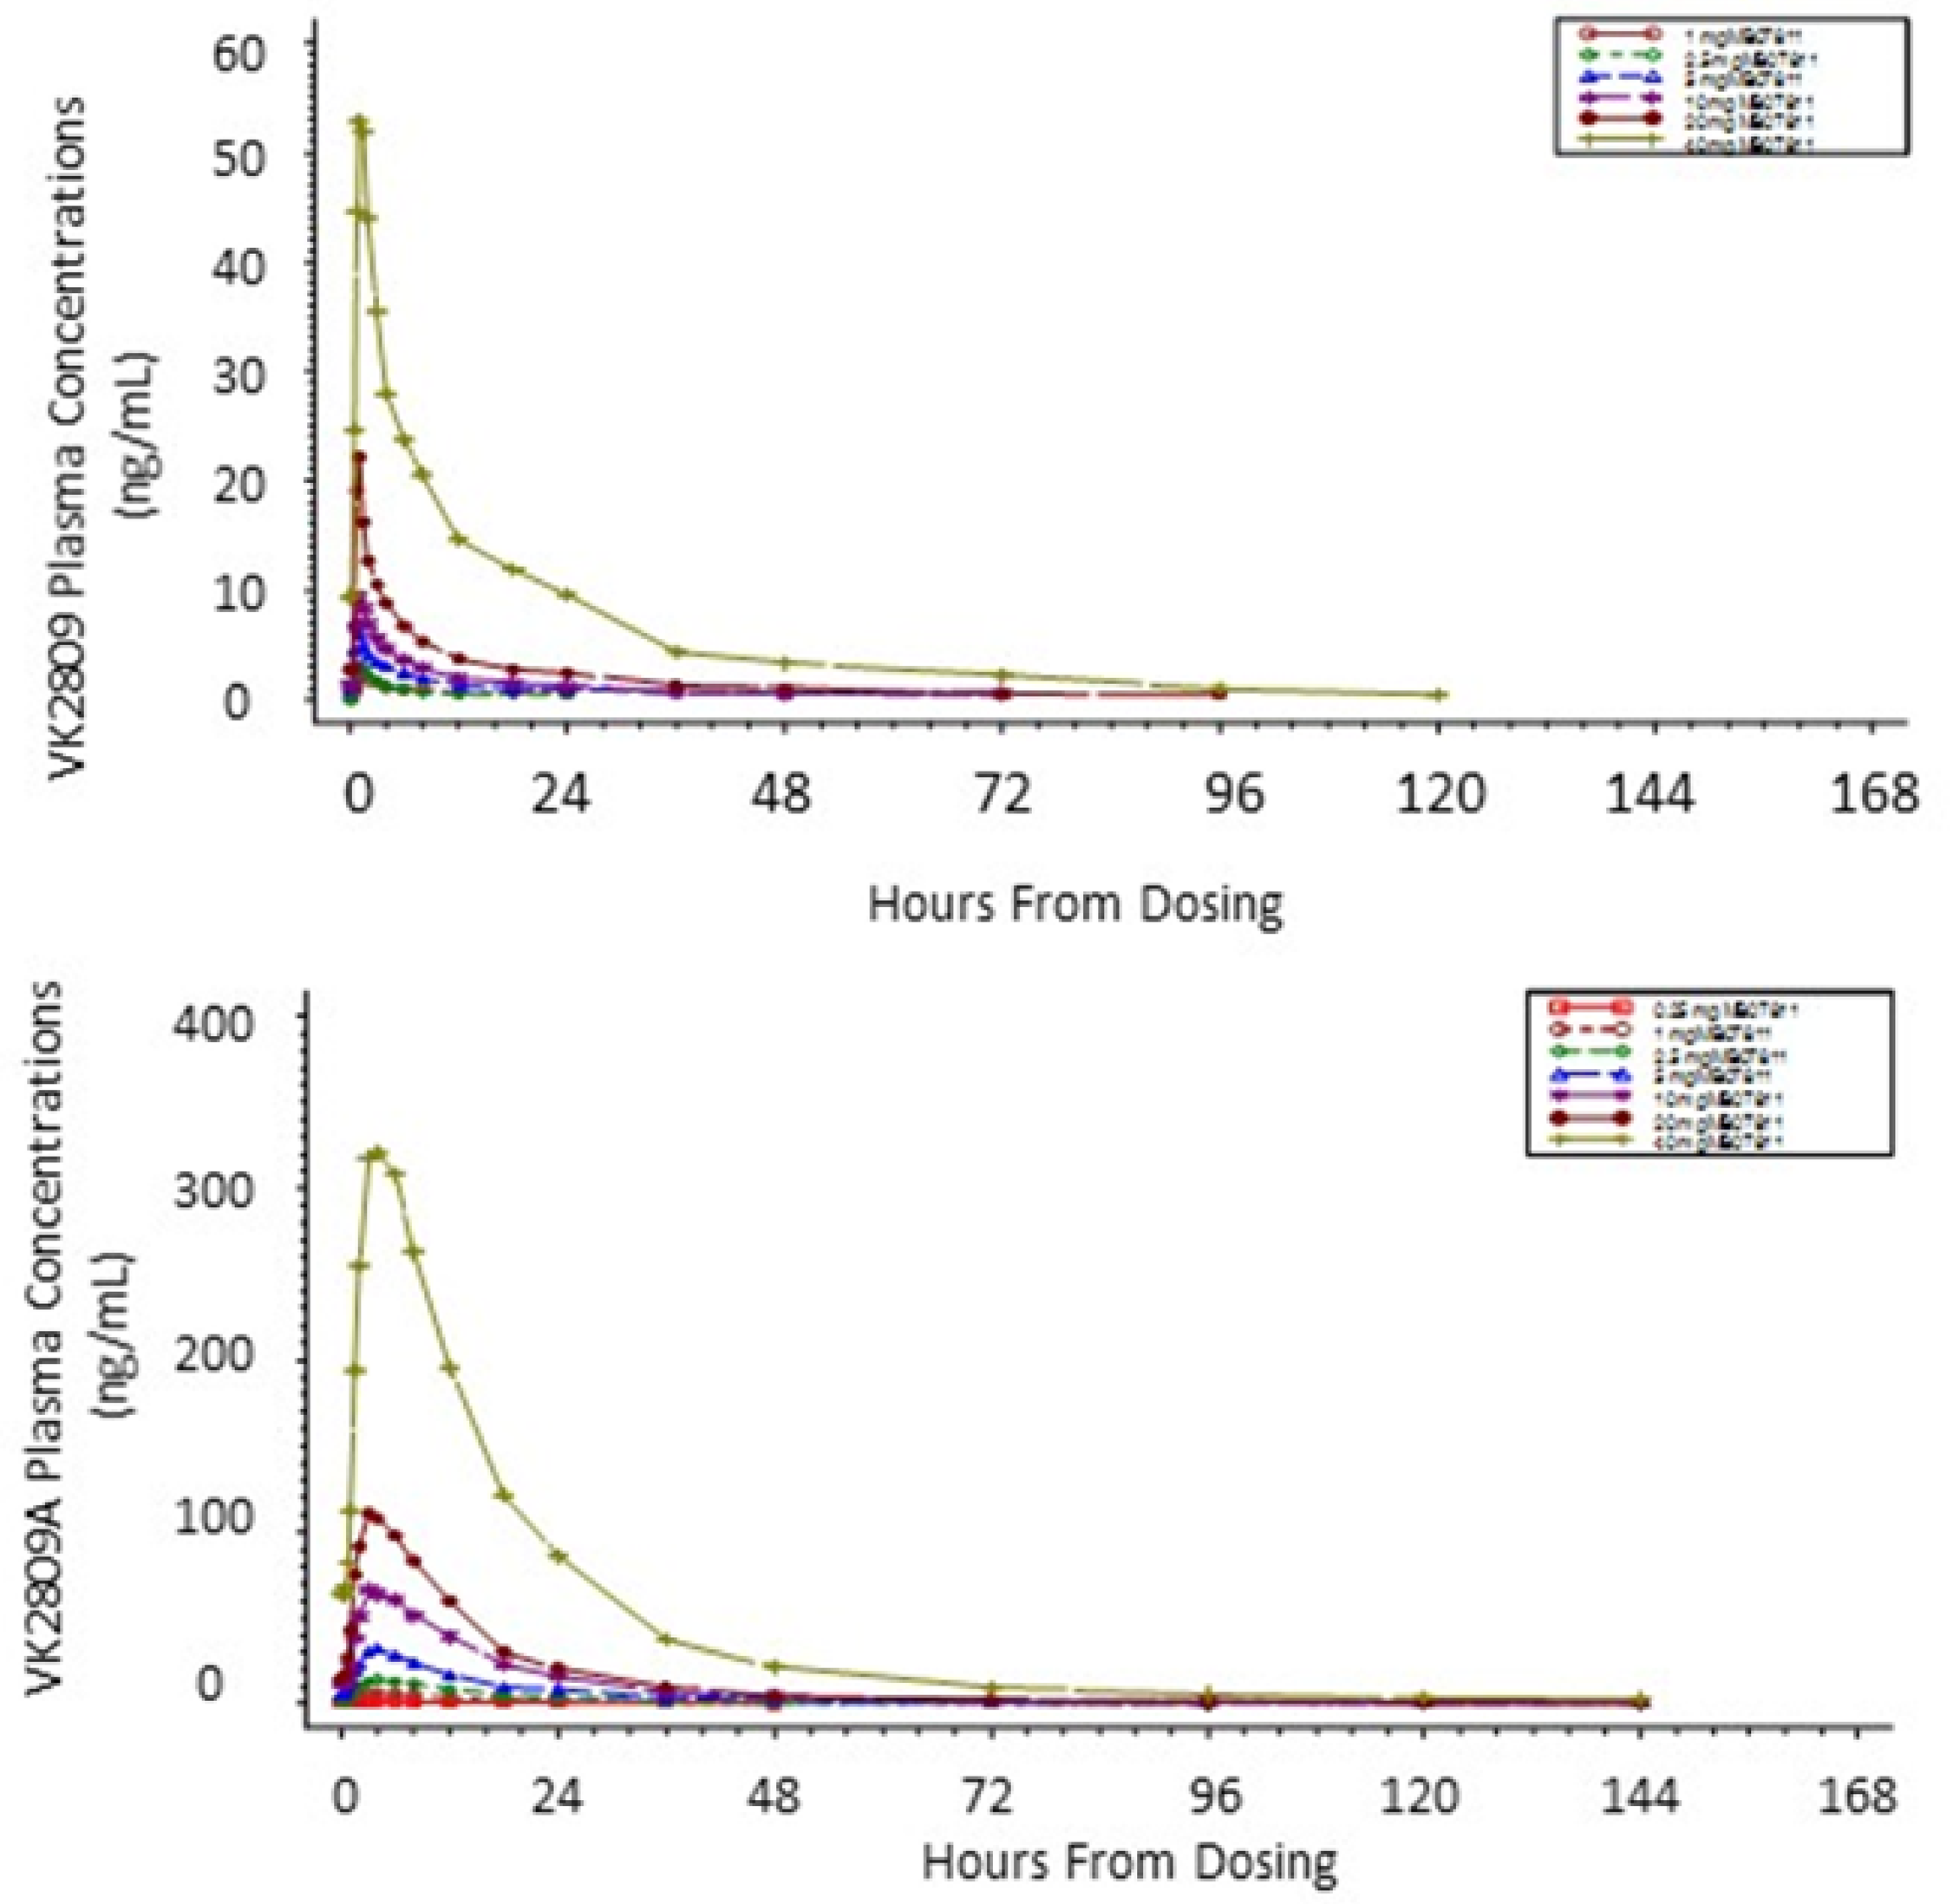

VK2809 Plasma Concentrations (ng/mL)
60
50
40
30
20
10
0
0 24 48 72 96 120 144 168
Hours From Dosing
VK2809A Plasma Concentrations (ng/mL)
400
300
200
100
0
0 24 48 72 96 120 144 168
Hours From Dosing

Effects on Lipids

Substantial and clinically meaningful placebo-adjusted reductions in LDL-C, up to 41.2% ($p<0.05$); triglycerides, up to 78.6% ($p<0.05$); and non-HDL-C, up to 44.2% ($p<0.05$), were observed at doses of 5.0 mg and above. In addition, significant reductions in Lp(a) and Apo-B were observed. No meaningful trends in HDL-C were observed.

Dose	Placebo-Adjusted LSM (SE)				
	LDL-C	Triglyceride	TC	HDL-C	Non-HDL-C
0.25 mg (N=6)	10.5 (6.6)	-9.6 (17.5)	4.0 (4.9)	2.3 (7.0)	6.0 (7.1)
1 mg (N=6)	-9.2 (6.6)	9.7 (17.1)	-2.4 (5.0)	-2.2 (6.9)	-2.6 (7.1)
2.5 mg (N=6)	-0.7 (6.6)	-36.6 (17.3)	-1.1 (4.9)	11.2 (6.9)	-4.5 (7.0)
5 mg (N=6)	-15.2 (6.6)	-34.8 (17.4)	-15.7 (4.9)	-4.1 (6.9)	-19.9 (7.0)
10 mg (N=5)	-27.1 (7.0)	-61.0 (18.5)	-24.2 (5.4)	-3.0 (7.5)	-32.0 (7.5)
20 mg (N=6)	-41.2 (6.6)	-62.1 (17.2)	-29.8 (5.0)	16.5 (6.9)	-44.2 (7.1)
40 mg (N=5)	-36.6 (7.0)	-78.6 (18.7)	-32.1 (5.3)	-2.7 (7.3)	-42.2 (7.5)

Unadjusted Change in LDL-C (Baseline to Day 15)

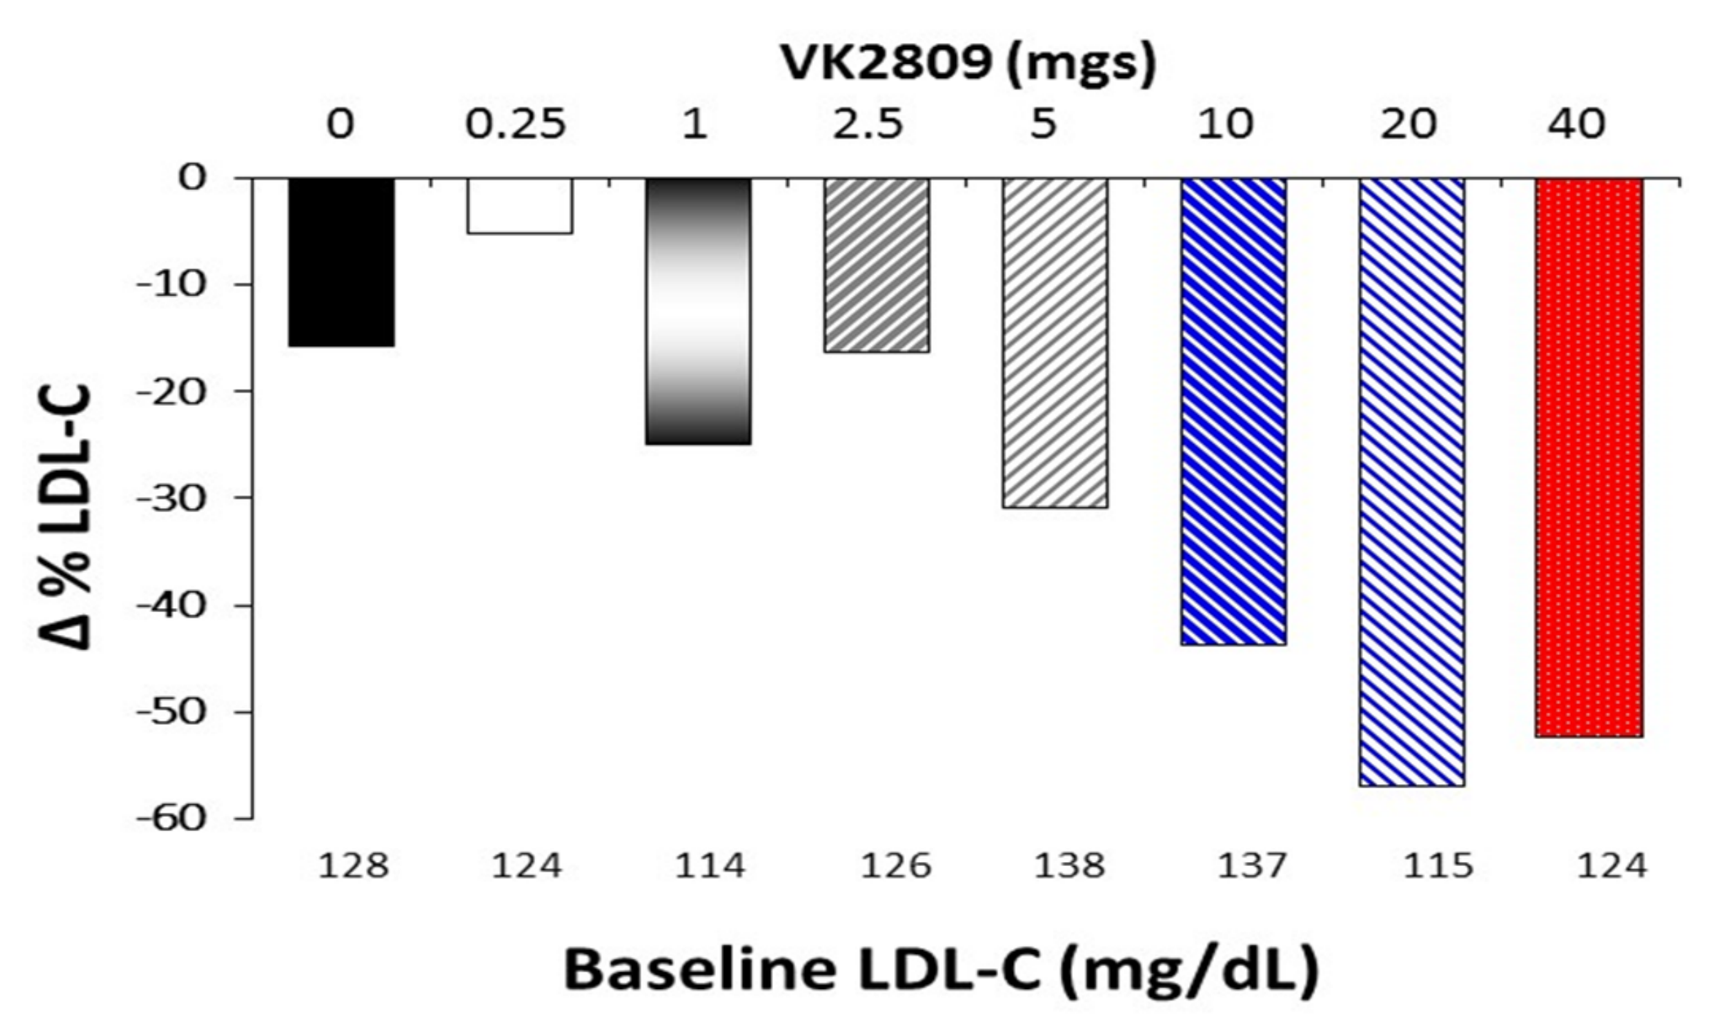

VK2809 (mgs)
0 0.25 1 2.5 5 10 20 40
Δ % LDL-C
0 -10 -20 -30 -40 -50 -60
128 124 114 126 138 137 115 124
Baseline LDL-C (mg/dL)

Unadjusted Change in Triglyceride (Baseline to Day 15)

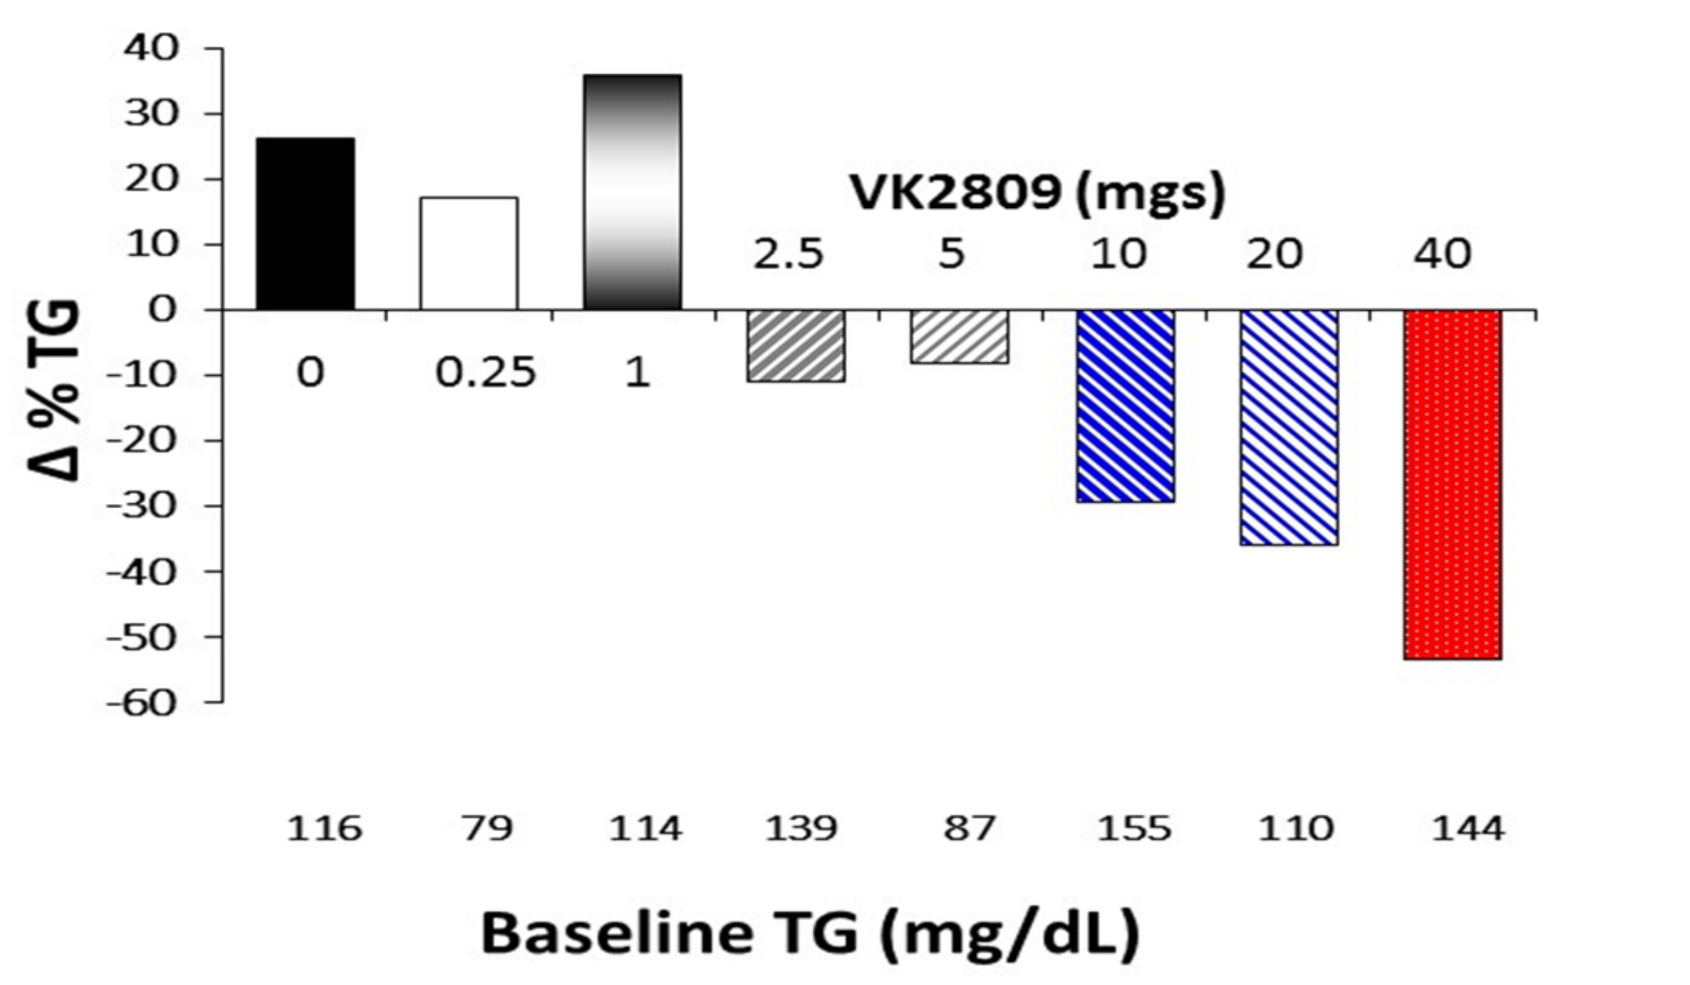

VK2809 (mgs)
0 0.25 1 2.5 5 10 20 40
Δ % TG
40 30 20 10 0 -10 -20 -30 -40 -50 -60
116 79 114 139 87 155 110 144
Baseline TG (mg/dL)

Unadjusted Change in APO B (Baseline to Day 15)

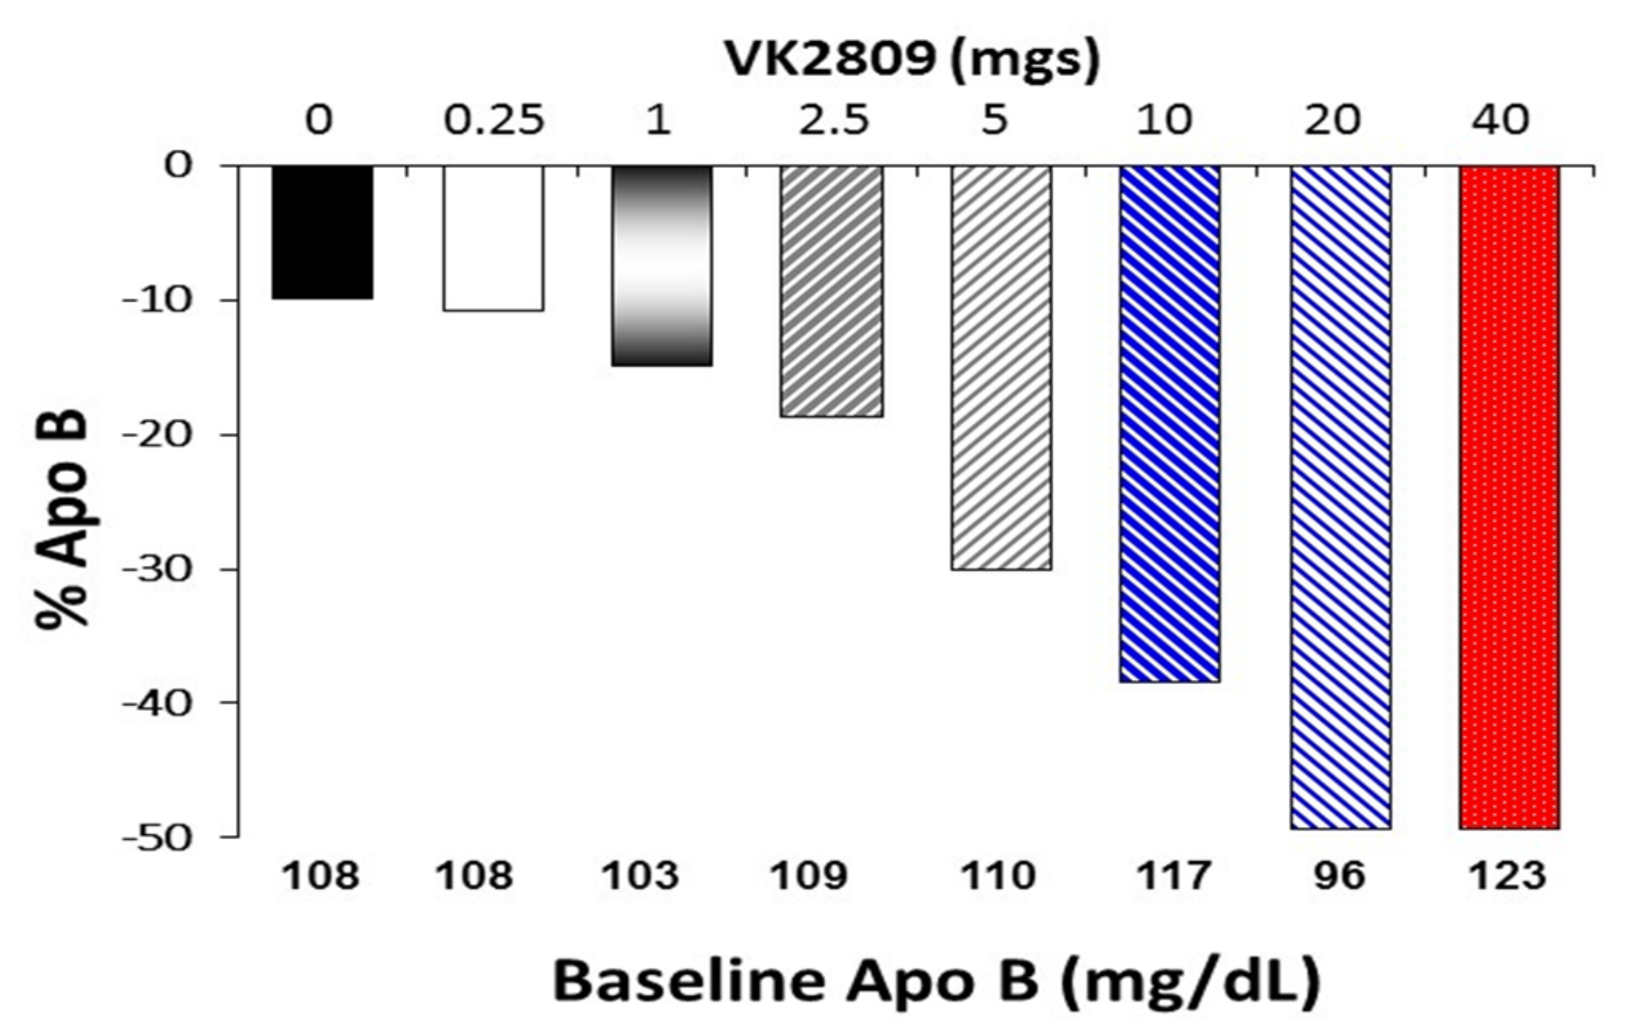

VK2809 (mgs)
0 0.25 1 2.5 5 10 20 40
% Apo B
0 -10 -20 -30 -40 -50
108 108 103 109 110 117 96 123
Baseline Apo B (mg/dL)

Unadjusted Change in Lp(a) (Baseline to Day 15)

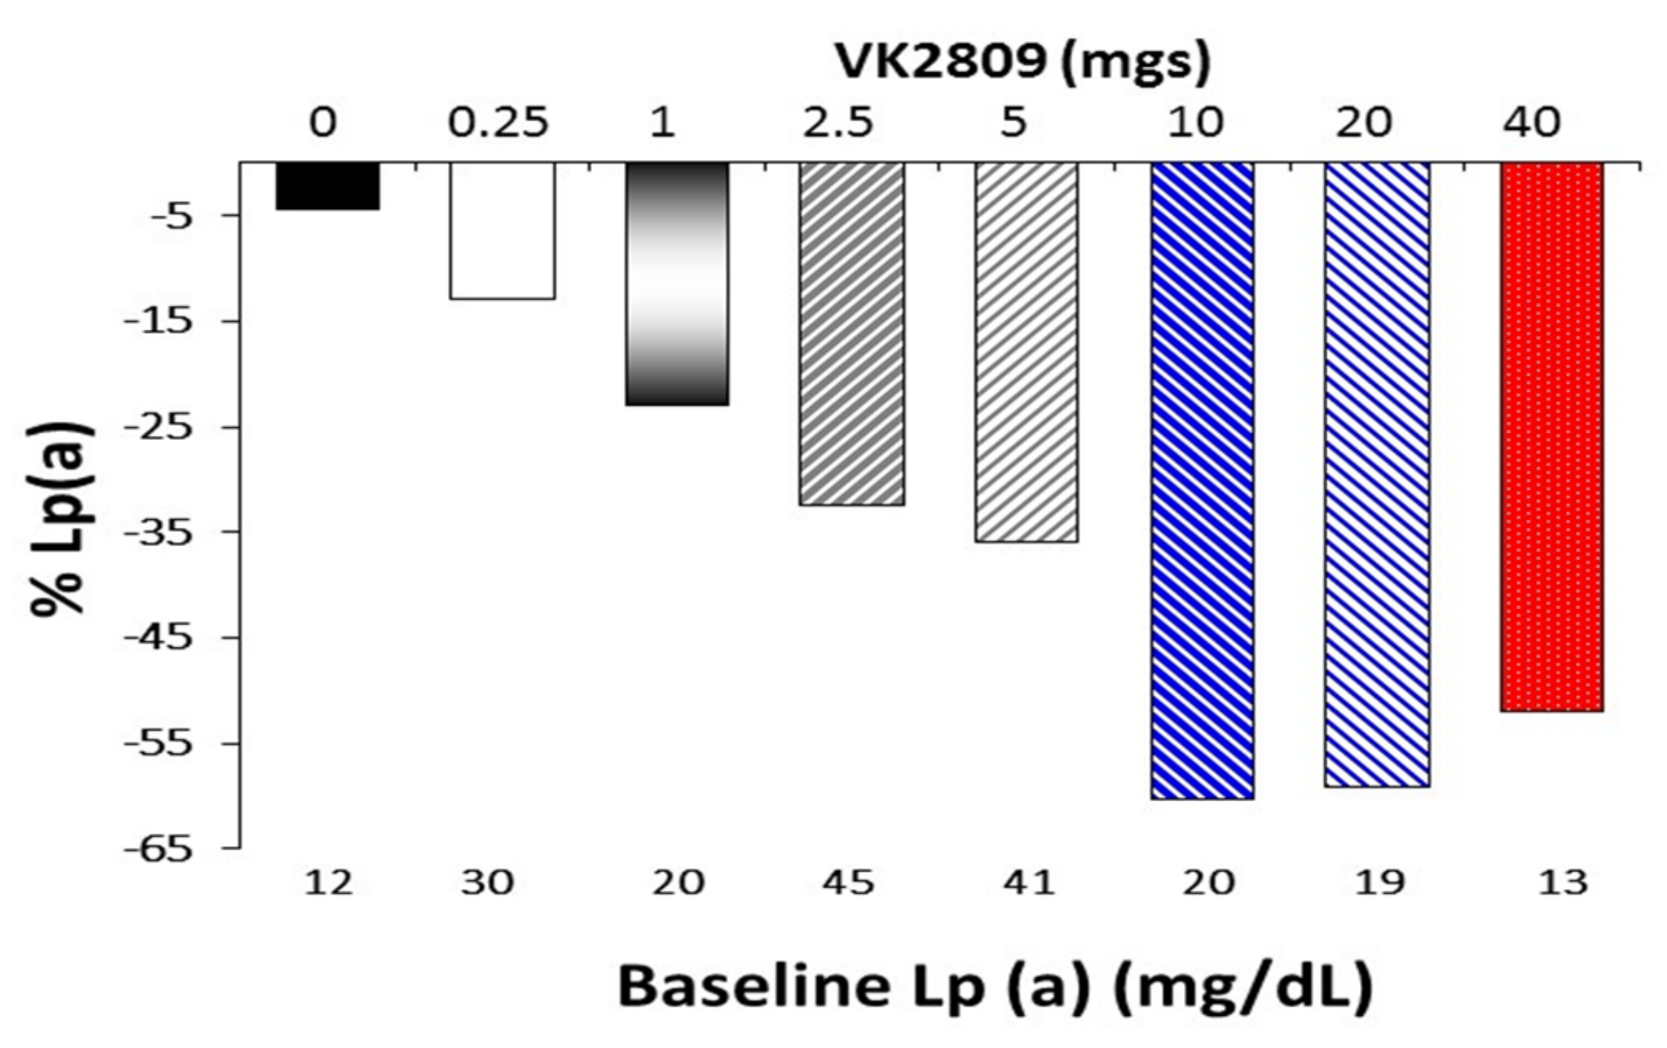

VK2809 (mgs)
0 0.25 1 2.5 5 10 20 40
% Lp(a)
-5 -15 -25 -35 -45 -55 -65
12 30 20 45 41 20 19 13
Baseline Lp (a) (mg/dL)

Safety

VK2809 was found to be safe and well-tolerated in this study. There were no serious adverse events. No treatment- or dose-related trends were observed for abnormal vital signs, physical examination assessments, ECGs or Holter-monitored cardiac rhythm. The frequency of AEs by dose was similar to placebo. Mean ALT levels reached a maximum of 1.5x ULN in the 40 mg cohort. Dose-dependent mean reductions in total and free T4, total T3 and TSH were observed beginning at day 8, but without symptoms or signs of thyroid dysfunction. No dose related or medically important trends were observed for fasting glucose, INR, or B-type natriuretic peptide.

Treatment-Emergent, Treatment-Related AEs in 2 or more VK2809-Treated Subjects in VK2809-102 (# of subjects)

	VK2809 DOSE GROUP (mg)							
	Placebo	0.25	1	2.5	5	10	20	40
Number of Subjects Dosed	14	6	6	6	6	6	6	6
ADVERSE EVENT								
Gastrointestinal Disorders								
Abdominal discomfort	0	0	0	0	0	0	1	1
General Disorders and Administration Site								
Fatigue	0	0	0	1	0	1	1	0
Investigations								
Alanine aminotransferase increased	0	0	1	1	1	1	3	3
Aspartate aminotransferase	0	0	0	0	0	1	1	2
Blood bilirubin increased	0	0	0	0	0	0	1	1
Blood bilirubin unconjugated	0	0	0	0	0	0	1	1
Urine colour abnormal	0	0	0	0	0	2	0	0
Nervous System Disorders								
Dizziness	0	0	0	1	0	0	0	1
Headache	4	0	0	1	0	1	2	2
Psychiatric disorders								
Insomnia	1	0	0	3	0	1	2	1

Data excerpted from Appendix 16.2.7.2

Summary

- Daily administration of VK2809 for 14 days was safe and well-tolerated by male and female subjects with elevated serum cholesterol in this study. Following multiple once-daily dosing with VK2809, the extent of exposure [AUC(0-tz) and AUC(0-tau),ss] to VK2809 appeared dose proportional within the 1 to 40 mg dose range. Dose proportionality could not be shown for Cmax,ss. Individual Tmax for VK2809 occurred around 0.5 - 3 hours. The mean apparent terminal t1/2 ranged from 7 - 20 hours, with shorter values at lower doses compared to higher doses.
- The peak and extent of exposure to the active metabolite VK2809A increased in proportion to increase in VK2809 dose. Individual Tmax ranged from 1.5 - 6 hours post-dose. The mean apparent terminal t1/2 ranged from 13 - 41 hours and tended to be shorter following lower doses when compared to higher doses.
- VK2809 was not eliminated intact via a renal pathway, and only a small percentage (<3%) of administered VK2809 dose (based on molar ratios) was eliminated renally as VK2809A.
- Substantial and clinically meaningful placebo-adjusted reductions in LDL-C, up to 41.2% ($p<0.05$); triglycerides, up to 78.6% ($p<0.05$); and non-HDL-C, up to 44.2% ($p<0.05$), were observed at doses of 5.0 mg and above. In addition, significant reductions in Lp(a) and Apo-B were observed. No meaningful trends in HDL-C were observed.
- Asymptomatic elevation of liver enzymes and decreased thyroid levels were observed with 10 mg and higher daily doses.

Conclusions

- Selective activation of thyroid beta with the small molecule VK2809, as shown in this 14-day study, represents an attractive approach to the treatment of hypercholesterolemia.
- In this 14-day study the observed LDL-lowering effects were comparable to those historically reported with statin-based therapies. In addition, the observed robust reduction in plasma triglycerides suggest potential application in treating other conditions such as fatty liver disease.
- A Phase 2 trial evaluating the use of VK2809 in patients with increased cholesterol and fatty liver disease is planned.

Disclosures

Brian Lian, PhD: Officer, Director, Trustee, or other fiduciary role – Viking Therapeutics

Rochelle Hanley: MD: Officer, Director, Trustee, or other fiduciary role – Viking Therapeutics

Steven Schoenfeld, MD: Salary – Viking Therapeutics